UNIVERSITY OF MINNESOTA DULUTH
NATURAL RESOURCES RESEARCH INSTITUTE
Michael J. Lalich, Director

ECONOMIC GEOLOGY POSTER SERIES
NRRI POSTER-2009-05
Project 783-1237

NATURAL RESOURCES RESEARCH INSTITUTE

Minnesota Taconite Workers Lung Health Partnership — University of Minnesota

Taconite-Derived Mineral Dust in Population Centers on Mesabi Iron Range: Tracking Mineral Fibers from Ore to Air

Tamara Diedrich, Devon Brecke, Megan Schreiber, Larry Zanko
Natural Resources Research Institute, University of Minnesota Duluth

OBJECTIVE: Characterize mineral dust produced by mining taconite in Northern Minnesota with emphasis on elongated mineral particles

In an effort to address long-standing questions regarding the impact of dust derived from mining taconite on human health, the University of Minnesota is conducting multiple complementary health-related studies, including an exposure assessment, epidemiology studies, and exposure characterization research. As one of these studies, NRRI is performing a detailed characterization of the dust that is produced from mining and processing Biwabik Iron Formation ore, with emphasis on any mineral fibers present and the elongated mineral particles that are produced from mining and processing activities.

METHODS

A three-year long, field-based survey of taconite-derived mineral dust present in the air of communities directly surrounding the taconite operations of the Mesabi Iron Range is being conducted. This study is an opportunity for community sampling to be done in conjunction with in-plant sampling; the same metrics and techniques can be used in both environments.

Sampling Strategy

Air Sampling Along the Mesabi Iron Range

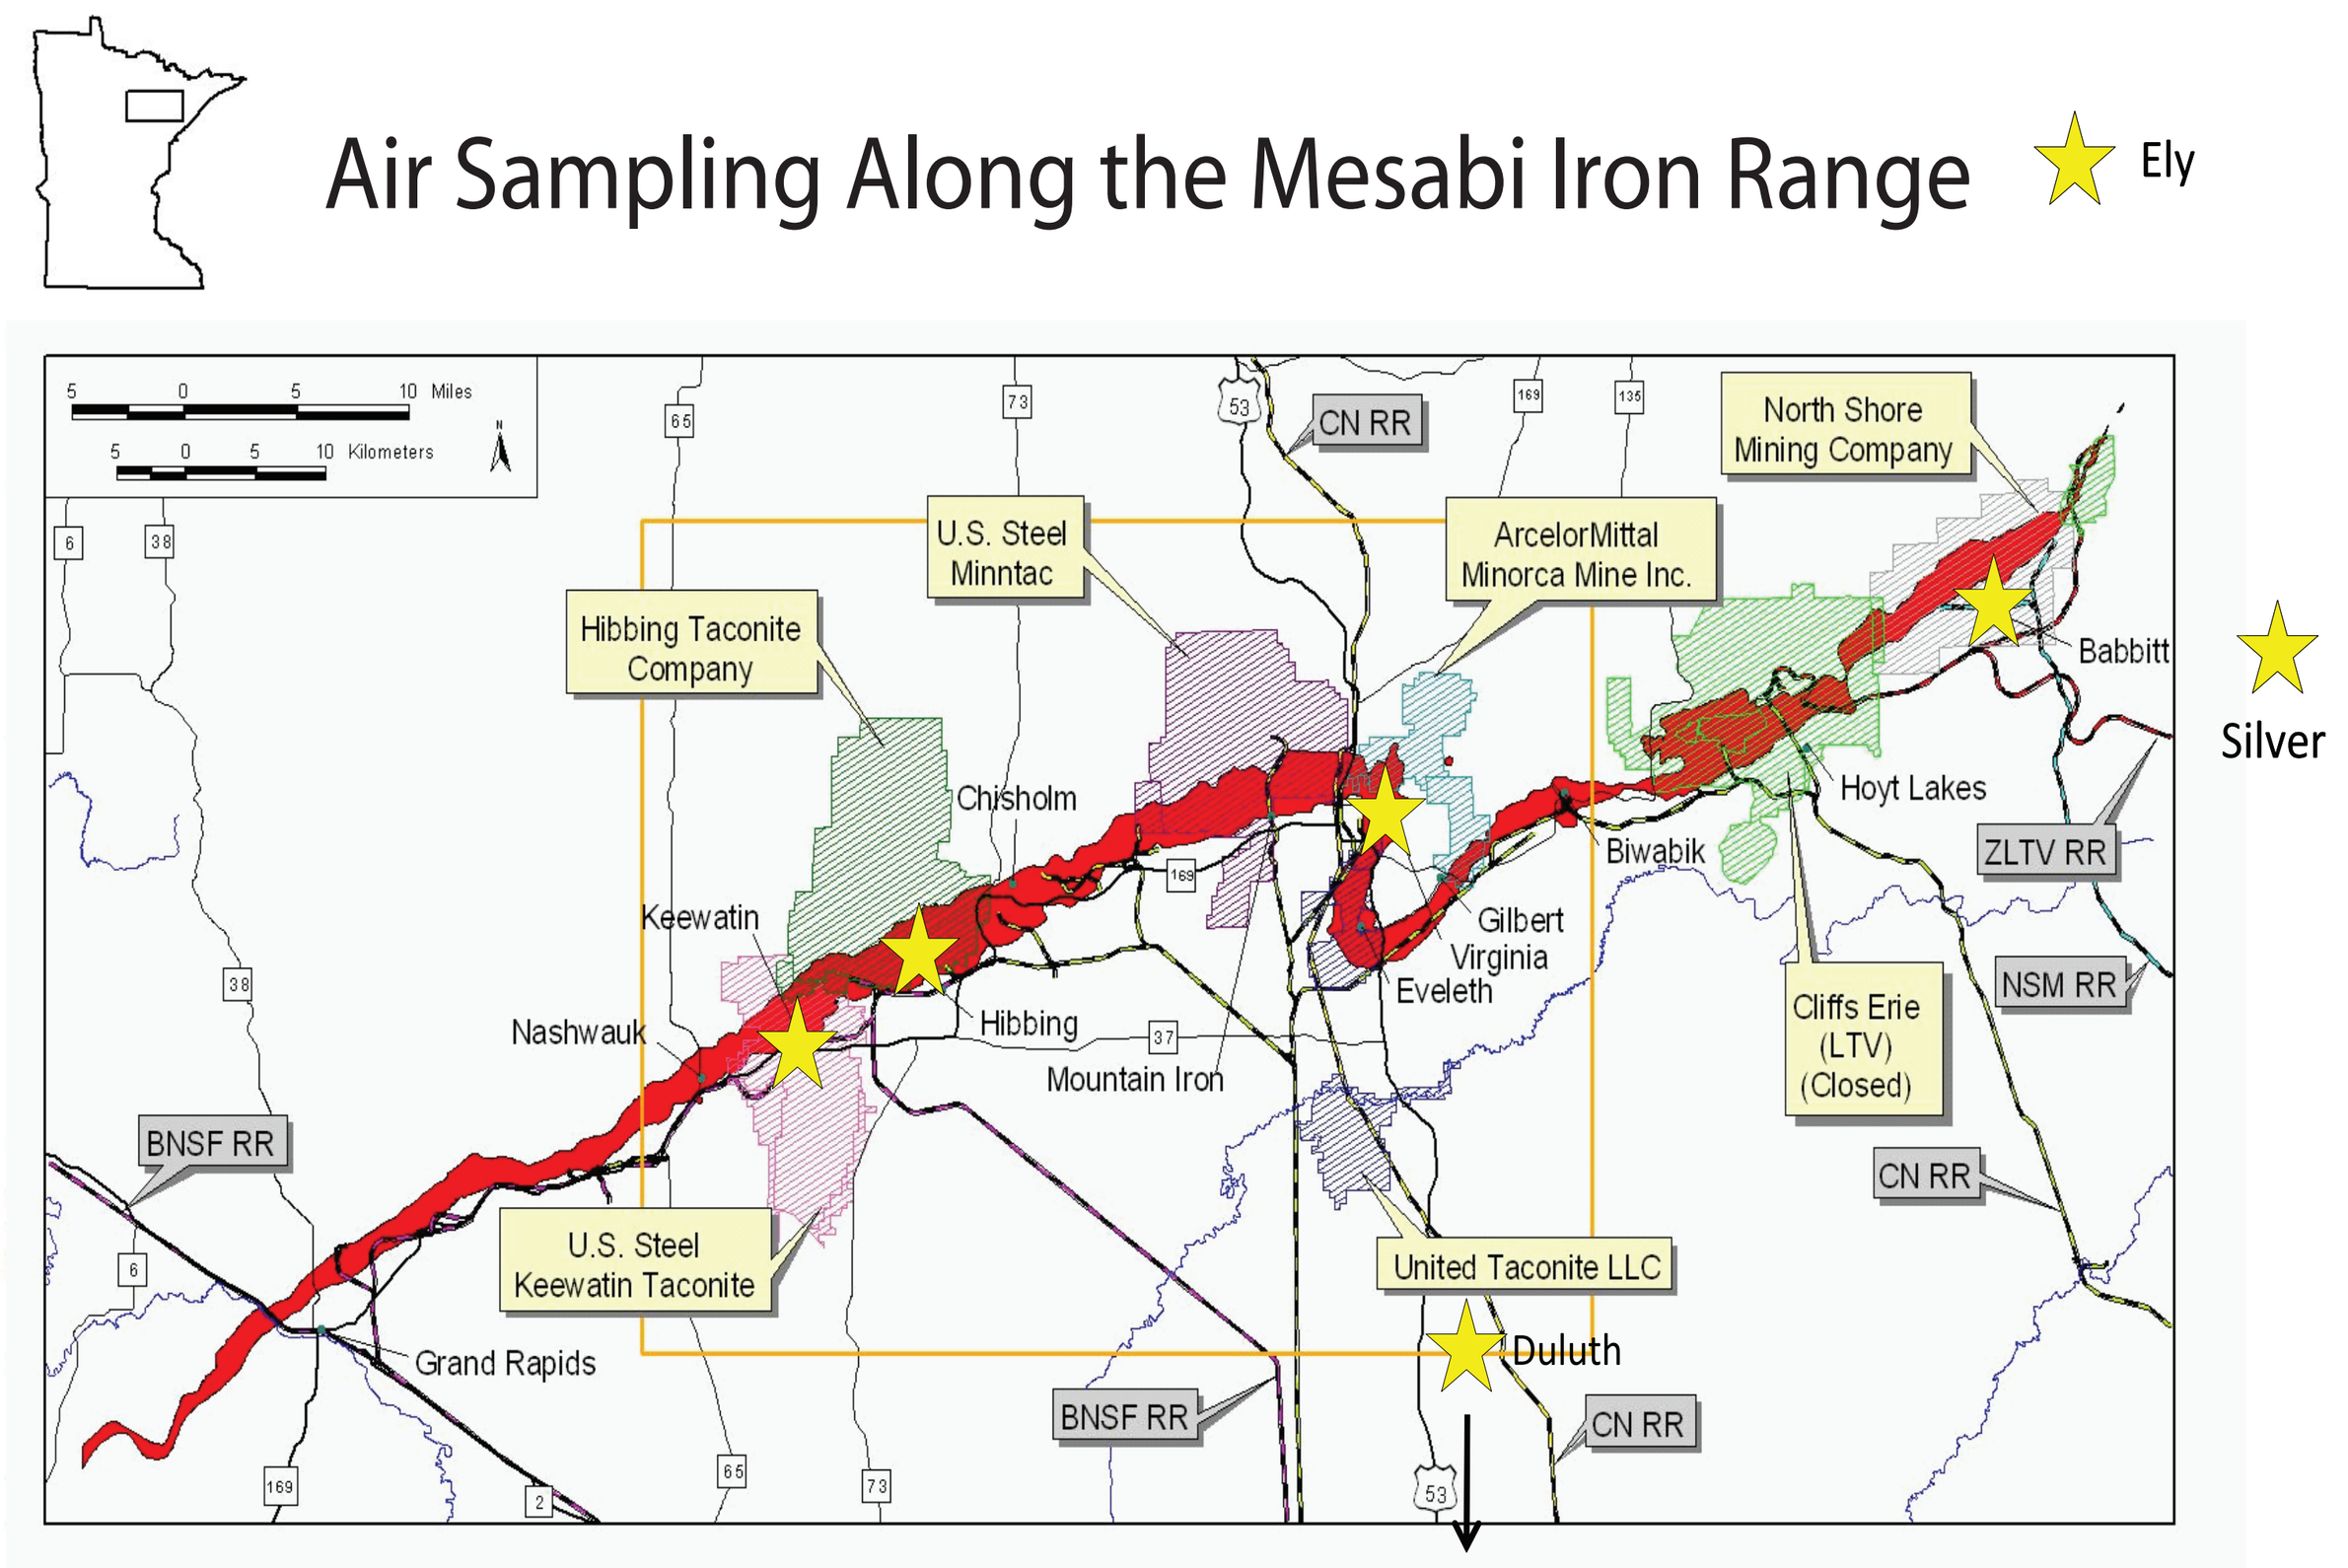
Ely
Silver Bay
Duluth
North Shore Mining Company
U.S. Steel Minntac
ArcelorMittal Minorca Mine Inc.
Hibbing Taconite Company
U.S. Steel Keewatin Taconite
United Taconite LLC
Cliffs Erie (LTV) (Closed)
Chisholm
Keewatin
Nashwauk
Hibbing
Mountain Iron
Gilbert
Virginia
Eveleth
Biwabik
Hoyt Lakes
Babbitt
Grand Rapids
CN RR
BNSF RR
ZLTV RR
NSM RR

Air sampling locations along Iron Range communities and processing plants (stars) as well as in Duluth and Ely for comparison. Samplers are placed in public locations: Keewatin Elementary School, Hibbing High School, Virginia City Hall, Babbitt Municipal Building, Ely Forest Service Station, Silver Bay High School, and Duluth NRRI.

Air Sampling Strategy

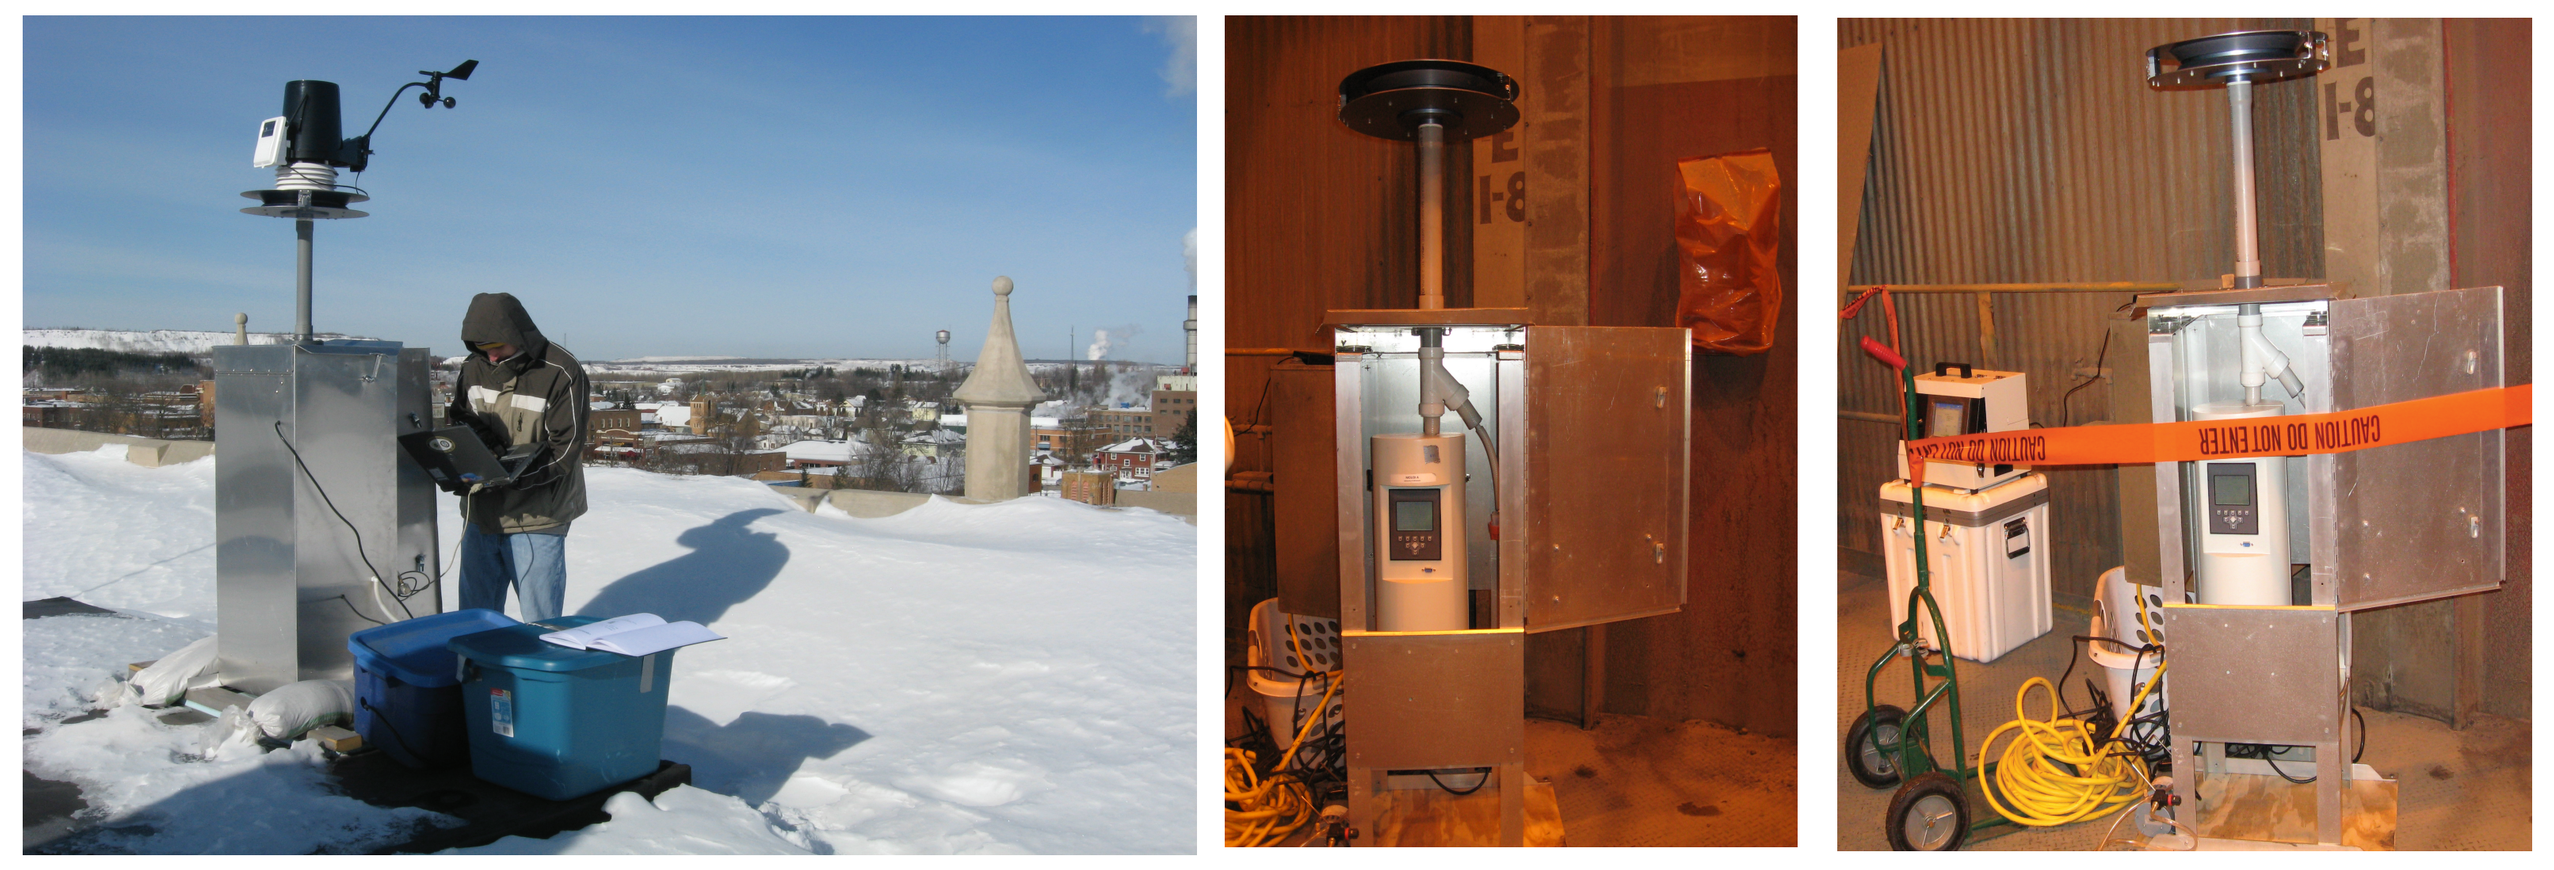

Air samples are collected using Micro-Orifice Uniform Deposit Impactors and are housed at sampling stations. Environmental example of impactor at Virginia Courthouse (left) and In-Plant example of impactor at Forbes, MN Pellet Plant (center and right).

Lake Sampling Strategy

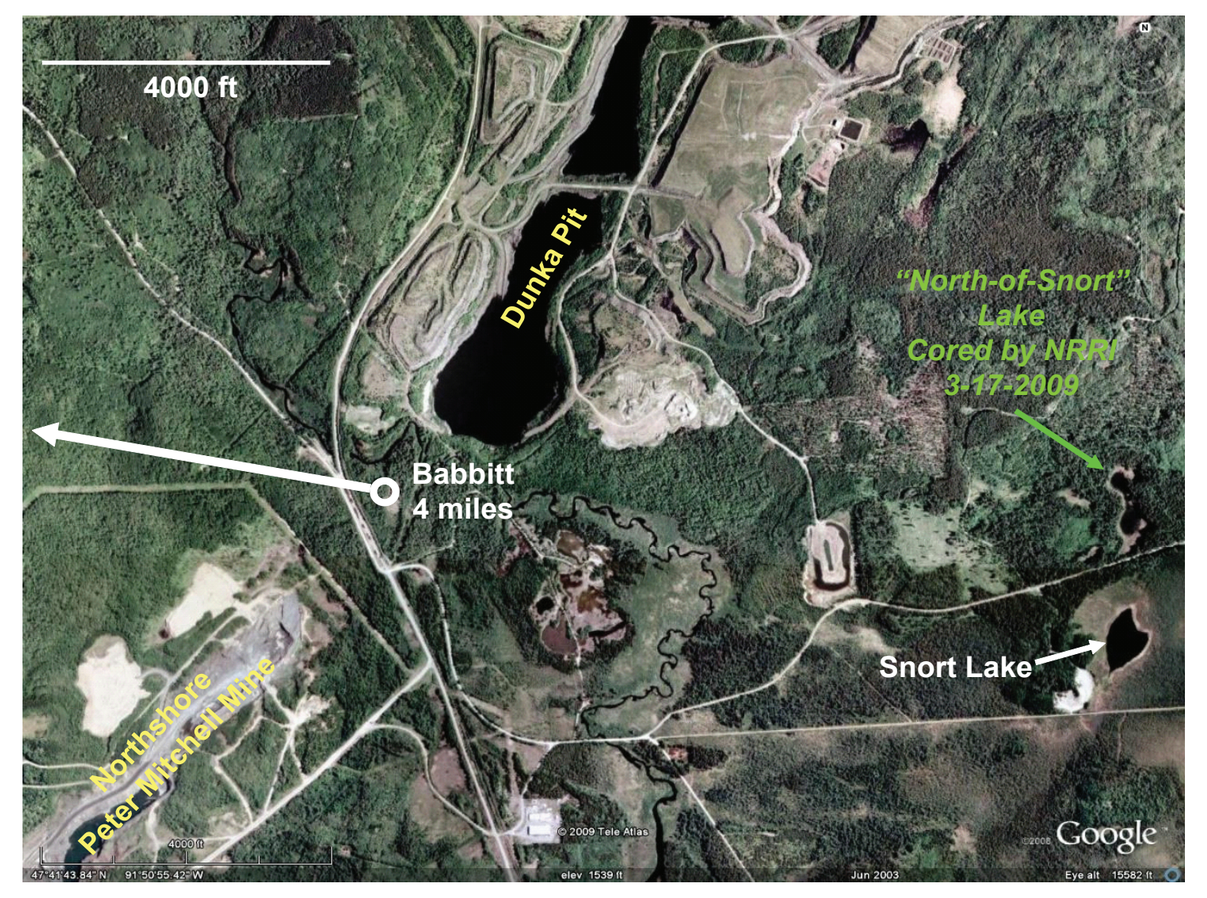
4000 ft
Dunka Pit
"North-of-Snort" Lake Cored by NRRI 3-17-2009
Babbitt 4 miles
Snort Lake
Northshore Peter Mitchell Mine
Google

Dated lake sediment cores from the west and east range will be used to evaluate how historic conditions compare with the present. "North-of-Snort" Lake (above) was cored in March, 2009.

Analytical Strategy

The characterization of mineral fibers and elongated mineral particles begins with the examination of fibrous minerals in situ from thin sections of metamorphosed and unmetamorphosed iron-formation. Additional analysis is completed on crushed material corresponding to these thin sections and the particulate matter present in the air of the taconite operations where the material is mined and processed. The analytical strategy for elongated mineral particles is to be as inclusive as possible with respect to what is being counted, while collecting detailed health-related data (minealogy, dimensions, chemistry).

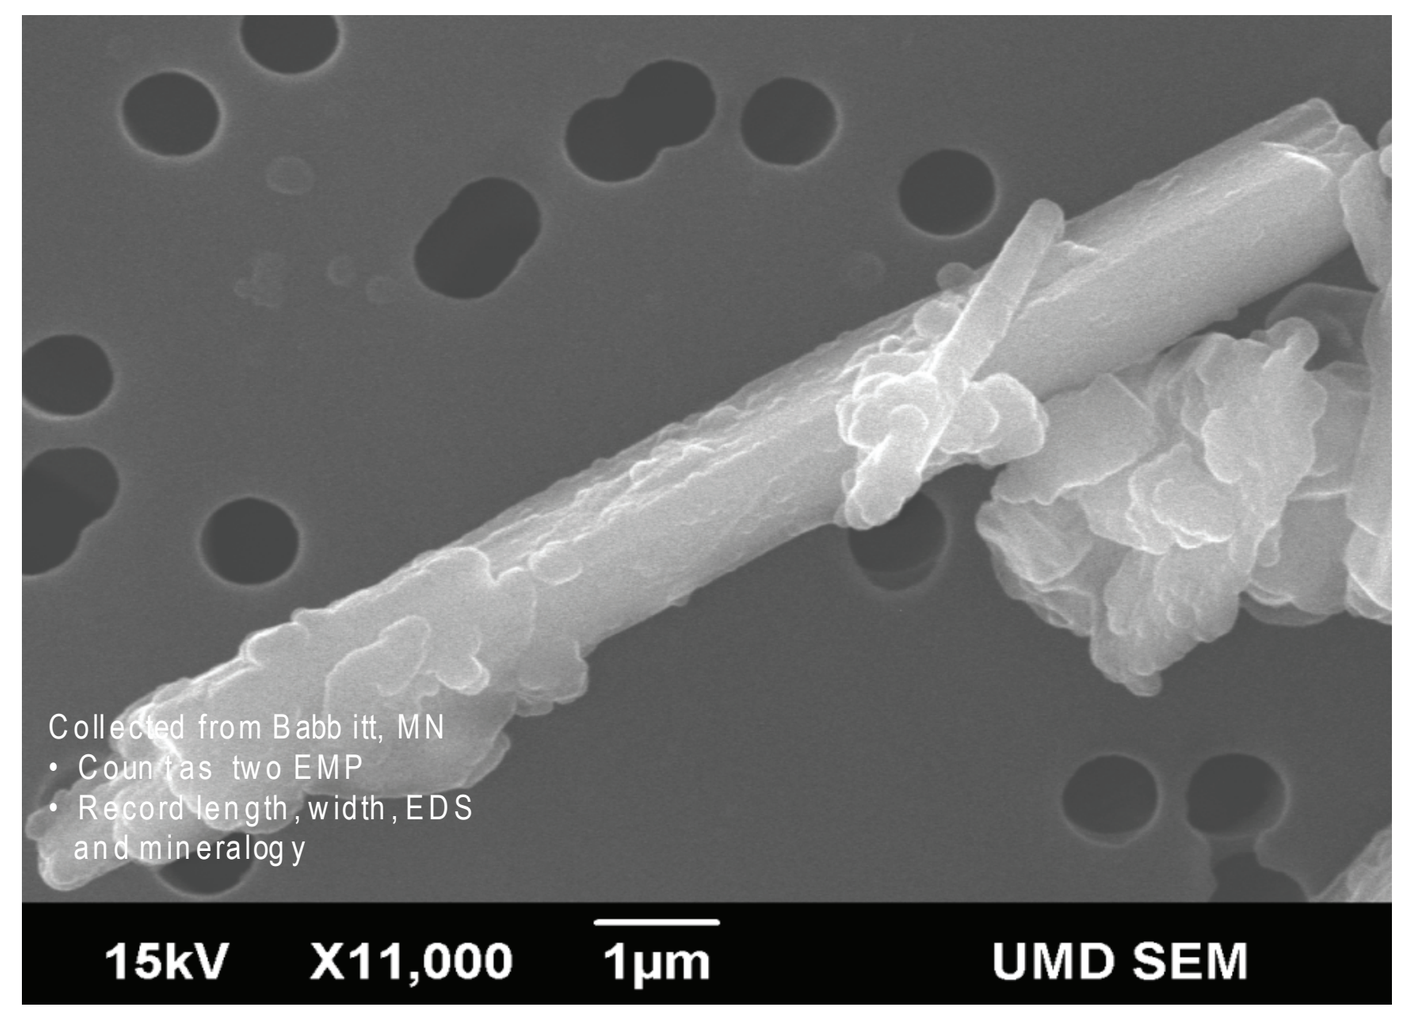
Collected from Babbitt, MN
• Counts as two EMP
• Record length, width, EDS and mineralogy
15kV X11,000 1µm UMD SEM

SEI SEM image of particle collected at Babbitt Municiple Building. Image includes brief procedure for EMP identification.

What is Asbestos?

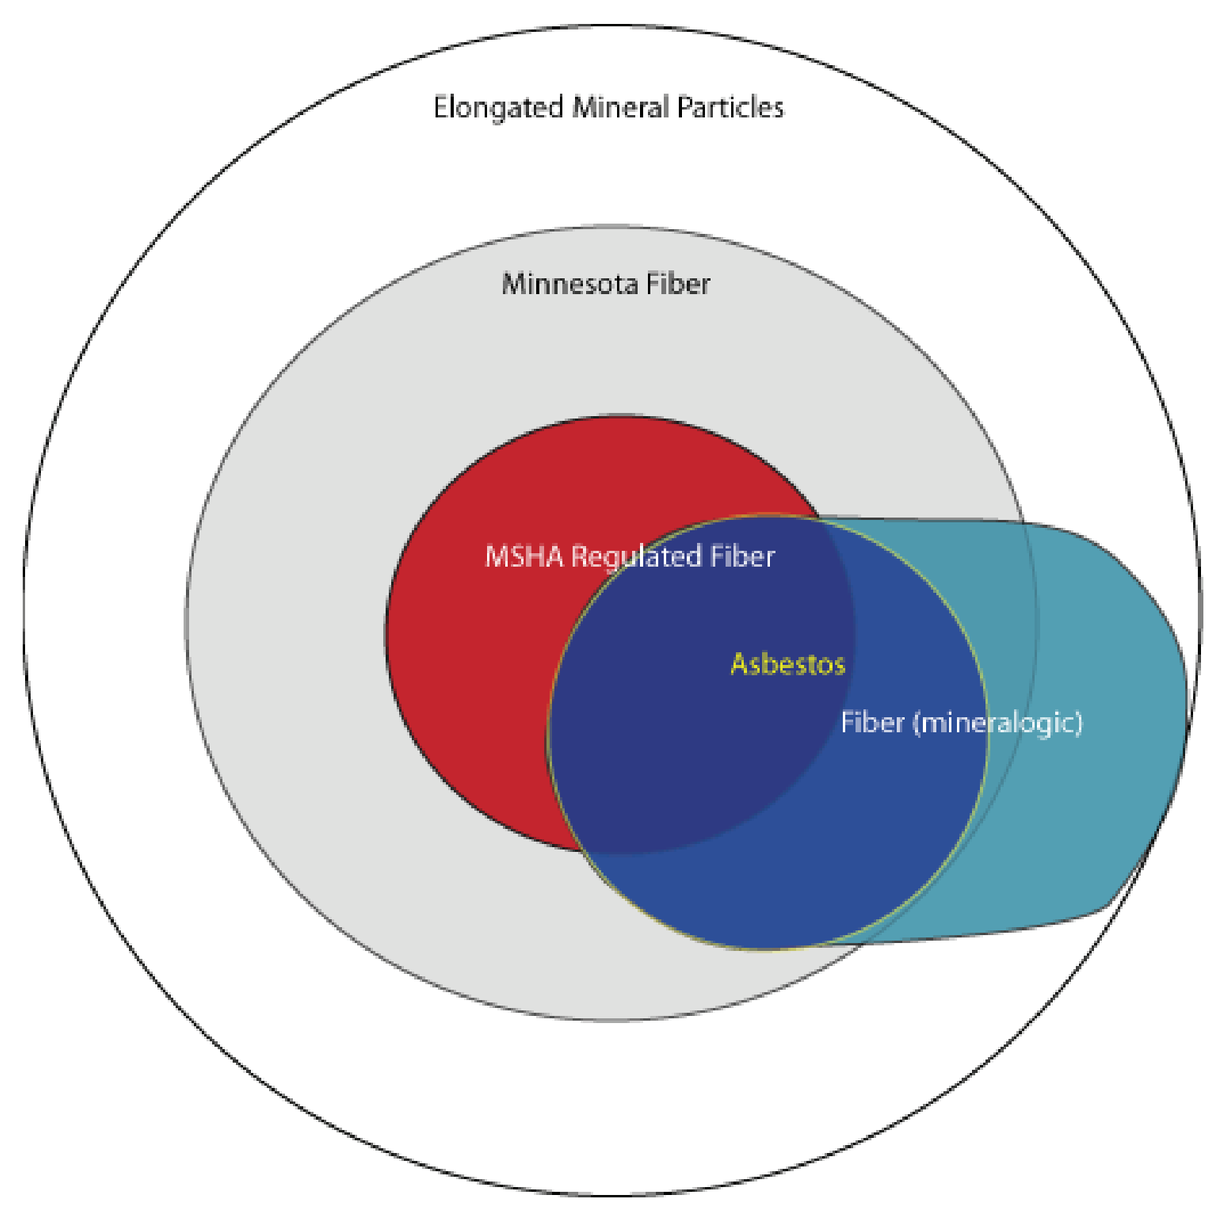
Elongated Mineral Particles
Minnesota Fiber
MSHA Regulated Fiber
Asbestos
Fiber (mineralogic)

Asbestos: Asbestiform variety of six minerals (chrysotile and five amphibole minerals.

Minnesota Fiber: Chrysotile and amphibole mineral particles with 3-to-1 or greater aspect ratios

MSHA-regulated Fiber: Chrysotile or regulated amphibole particle with an aspect ratio of 3:1 or greater, length of greater than 5 microns and a width greater than 0.25 microns.

Fiber: Crystal or polycrystalline aggregate displaying "fibrous" growth habit

Elongated Mineral Particle (EMP): "single crystal or similarly elongated polycrystalline aggregate particle with a length to width ratio of 3 to 1 or greater" (Bruce Vento Ban Asbestos and Prevent Mesothelioma Act of 2007)

PRELIMINARY DATA FROM VIRGINIA HORN AREA

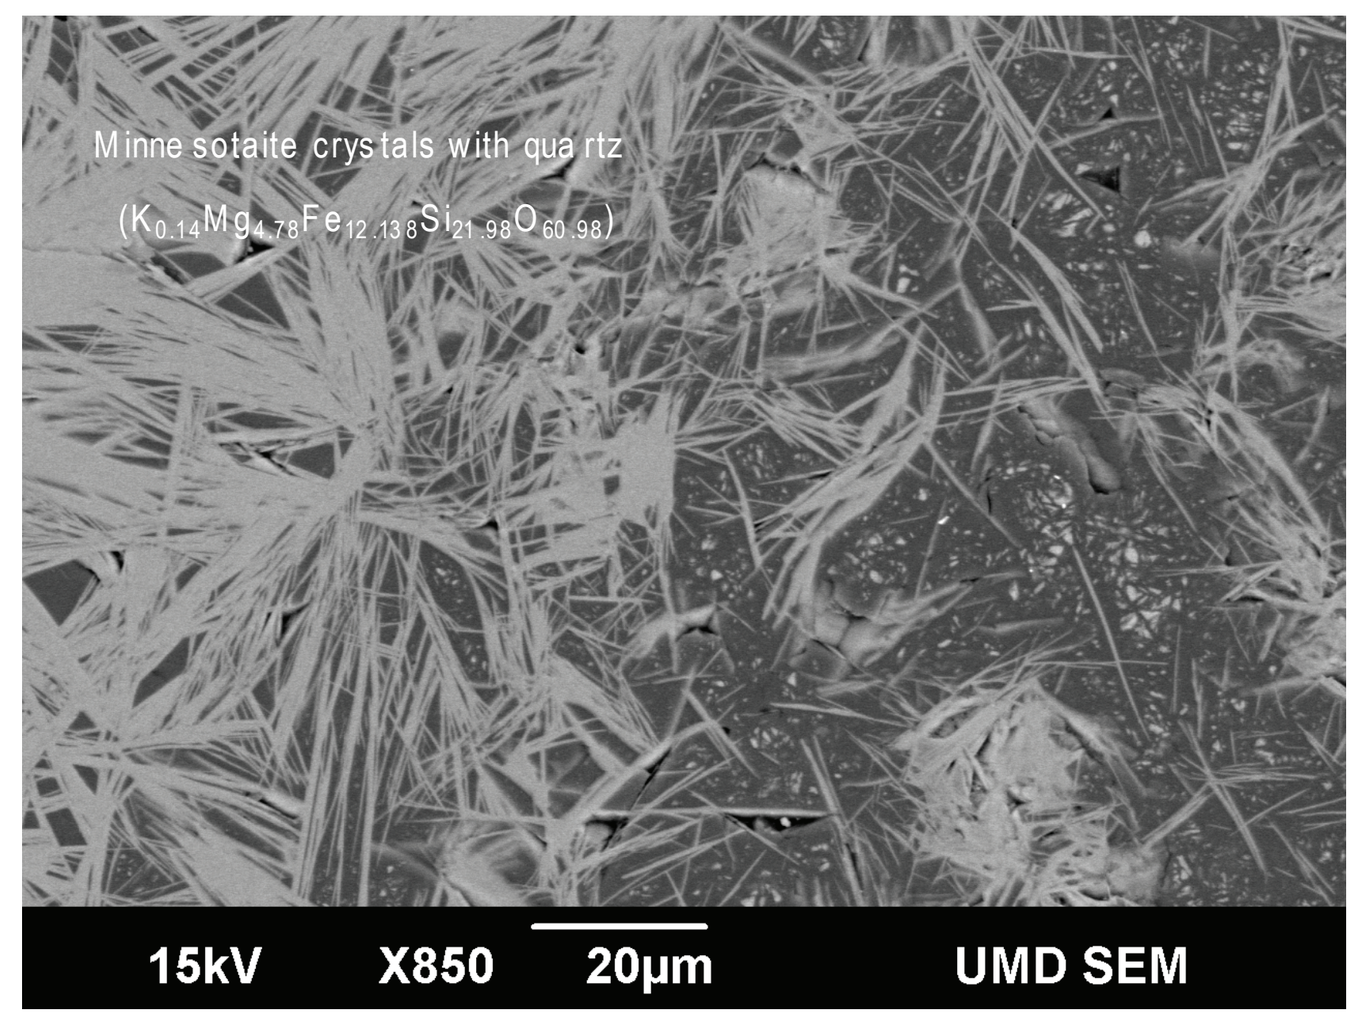
Minnesotaite crystals with quartz
15kV X850 20µm UMD SEM

BEI SEM image of elongated minnesotaite crystals (light grey) from a polished thin section of unmetamorphosed Biwabik Iron Formation. Minnesotaite is not an asbestos mineral, but does form elongated crystals.

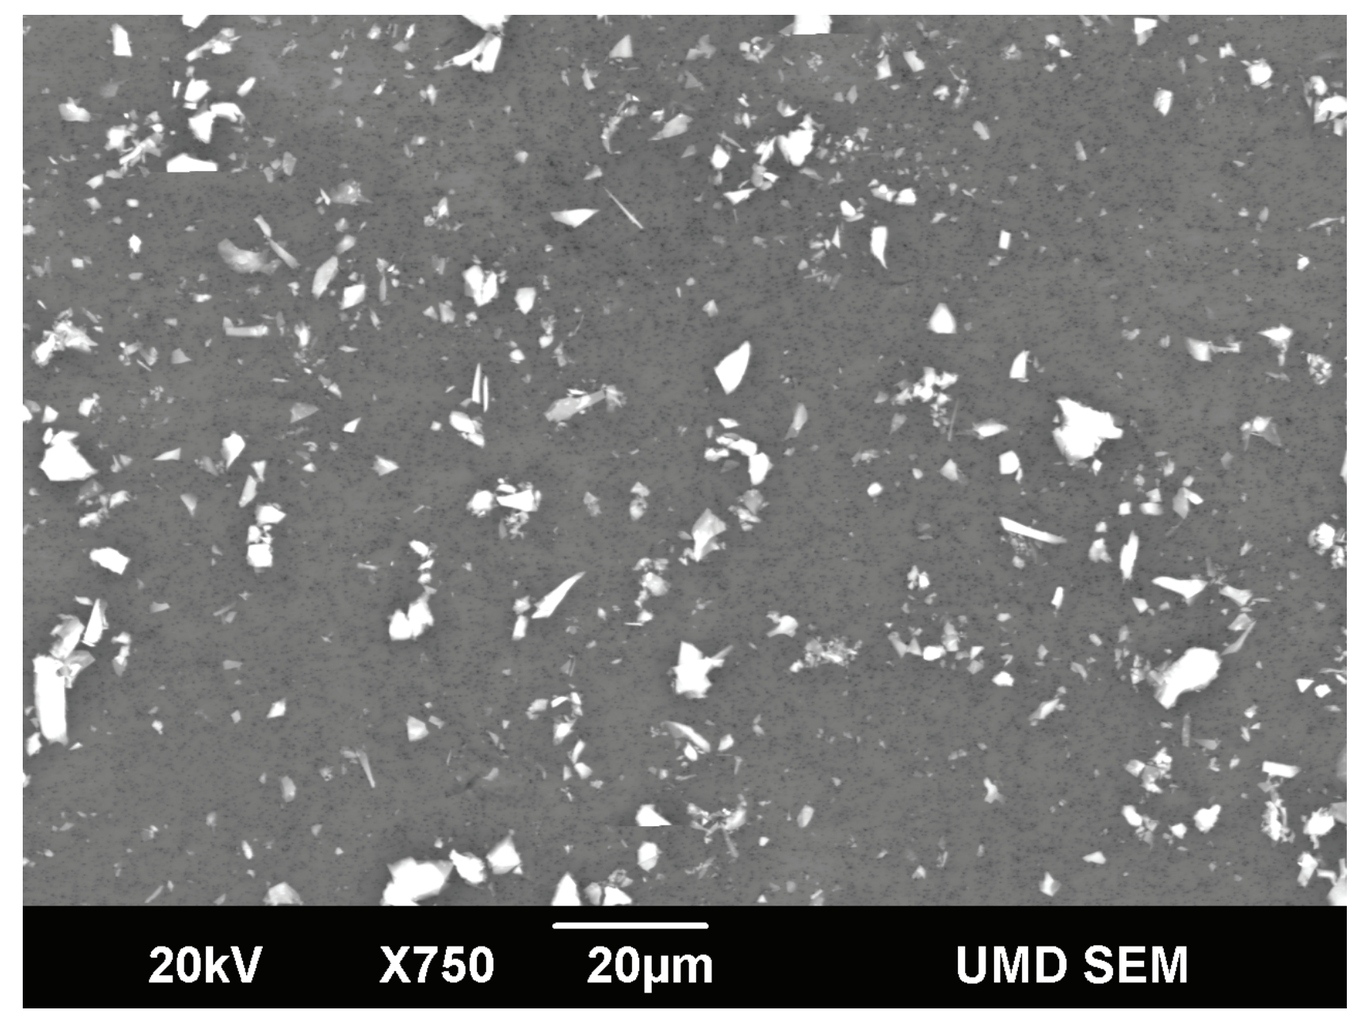
20kV X750 20µm UMD SEM

BEI SEM image of crushed rock corrosponding to polished thin section shown on the left.

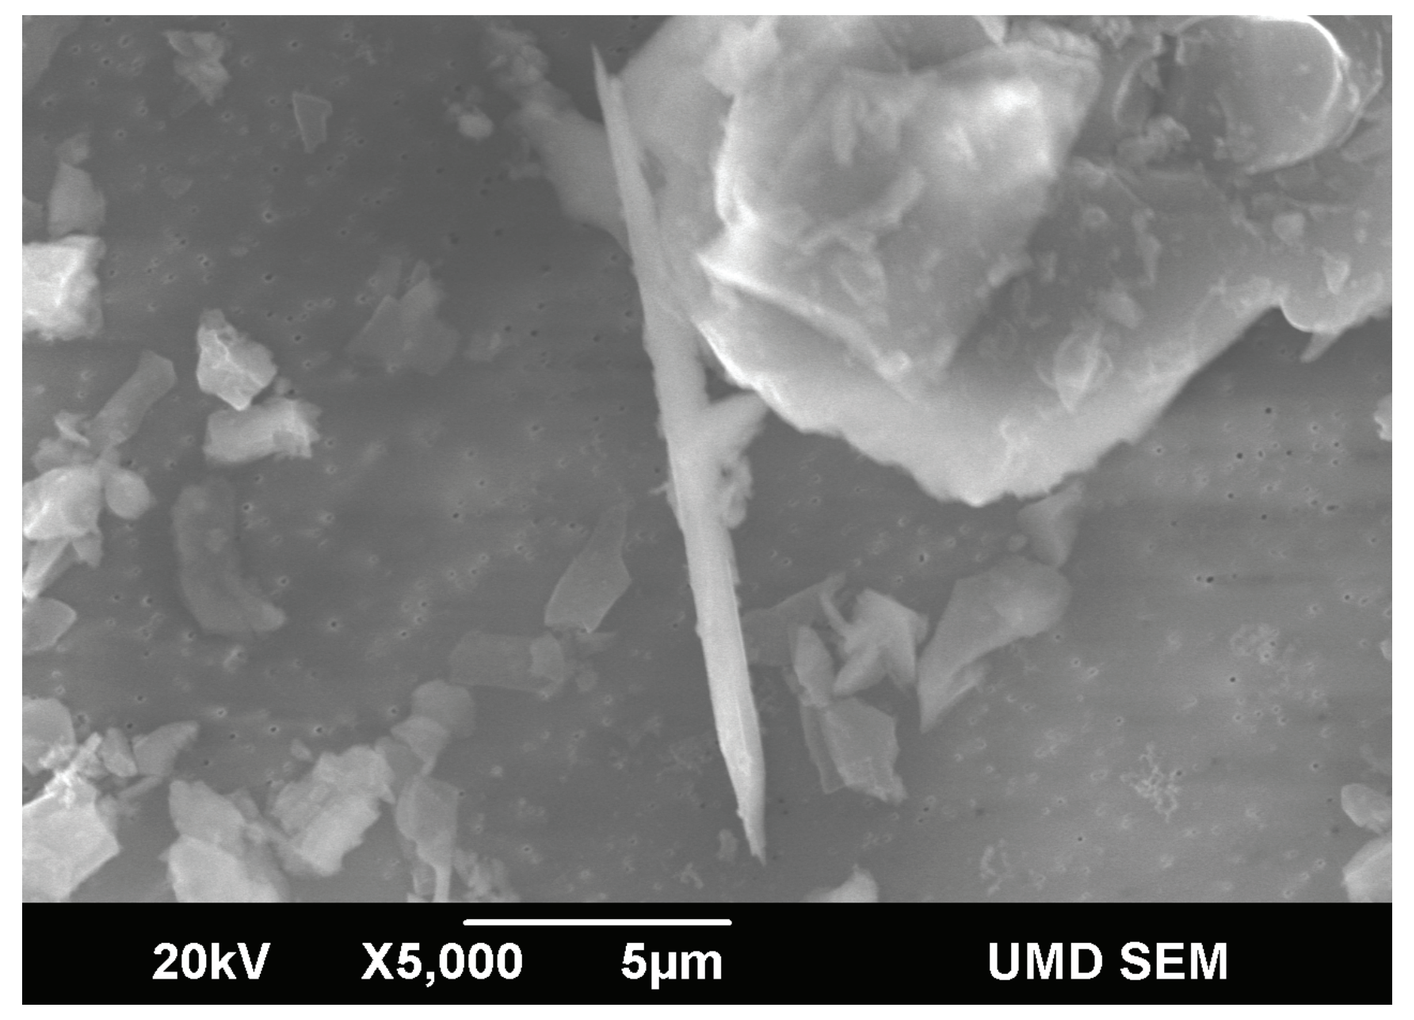
20kV X5,000 5µm UMD SEM

SEI SEM image of Minnesotaite particle liberated from crushing of the rock displayed in thin section.

In plant gravimetric data

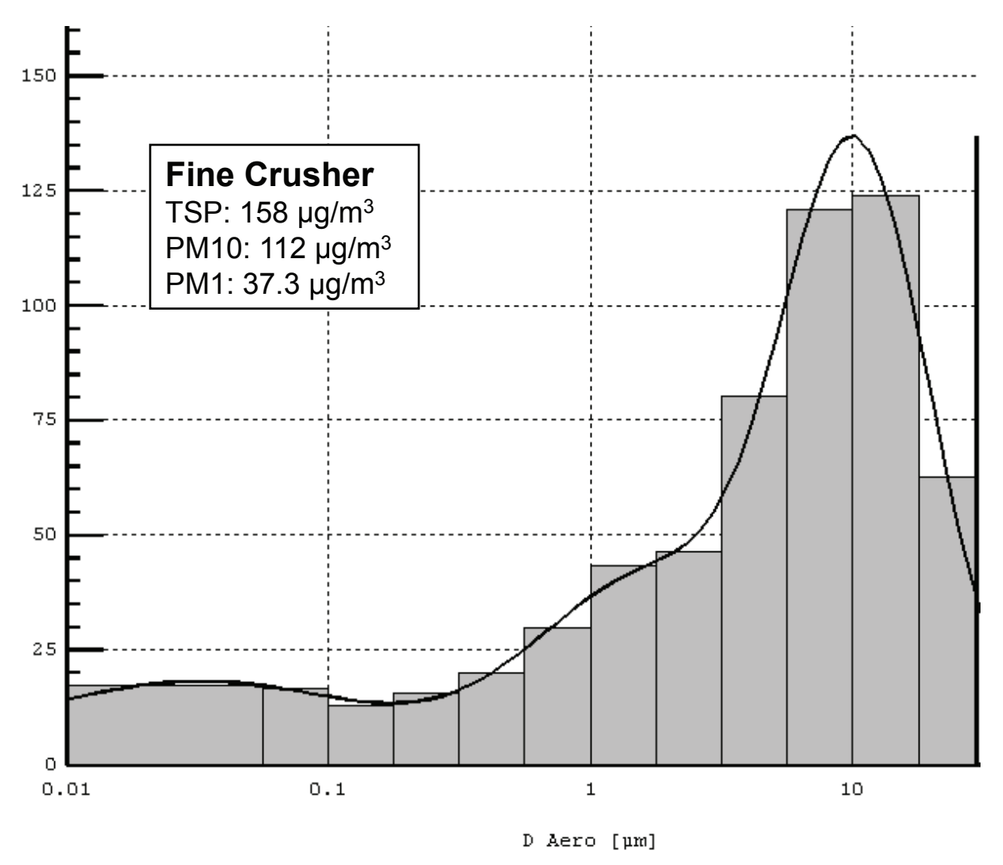
Fine Crusher
TSP: 158 µg/m3
PM10: 112 µg/m3
PM1: 37.3 µg/m3
D Aero [µm]

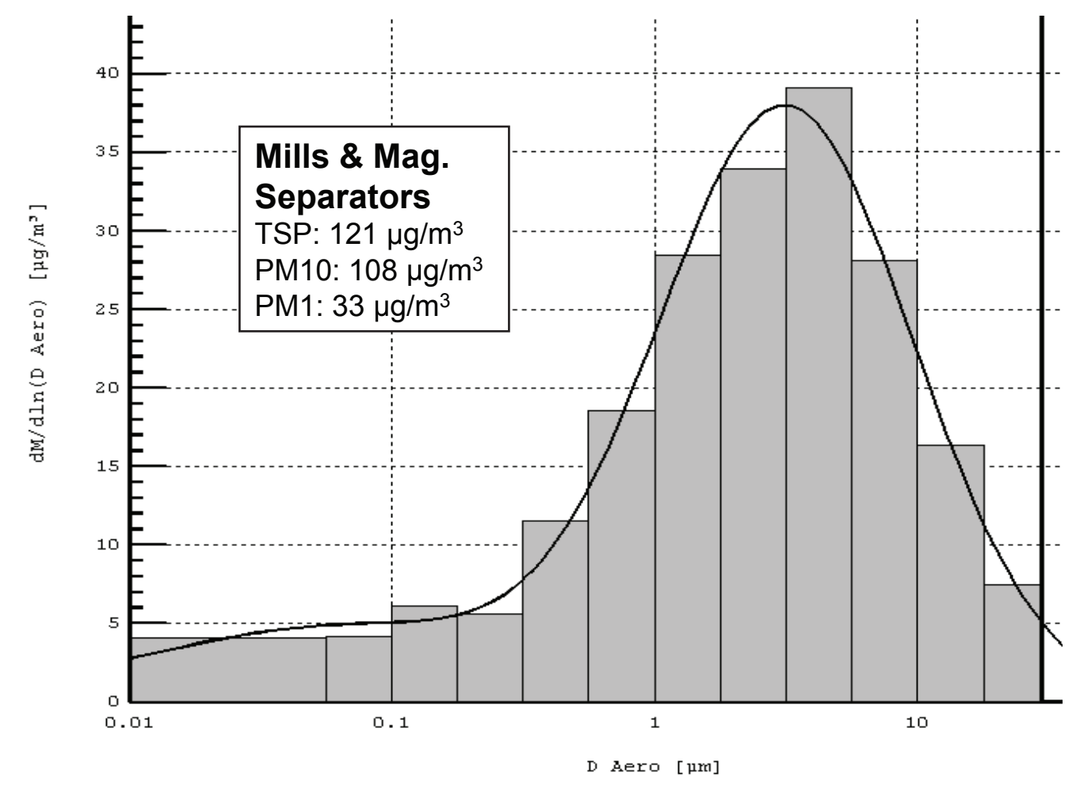
Mills & Mag. Separators
TSP: 121 µg/m3
PM10: 108 µg/m3
PM1: 33 µg/m3
dM/dln(D Aero) [µg/m3]
D Aero [µm]

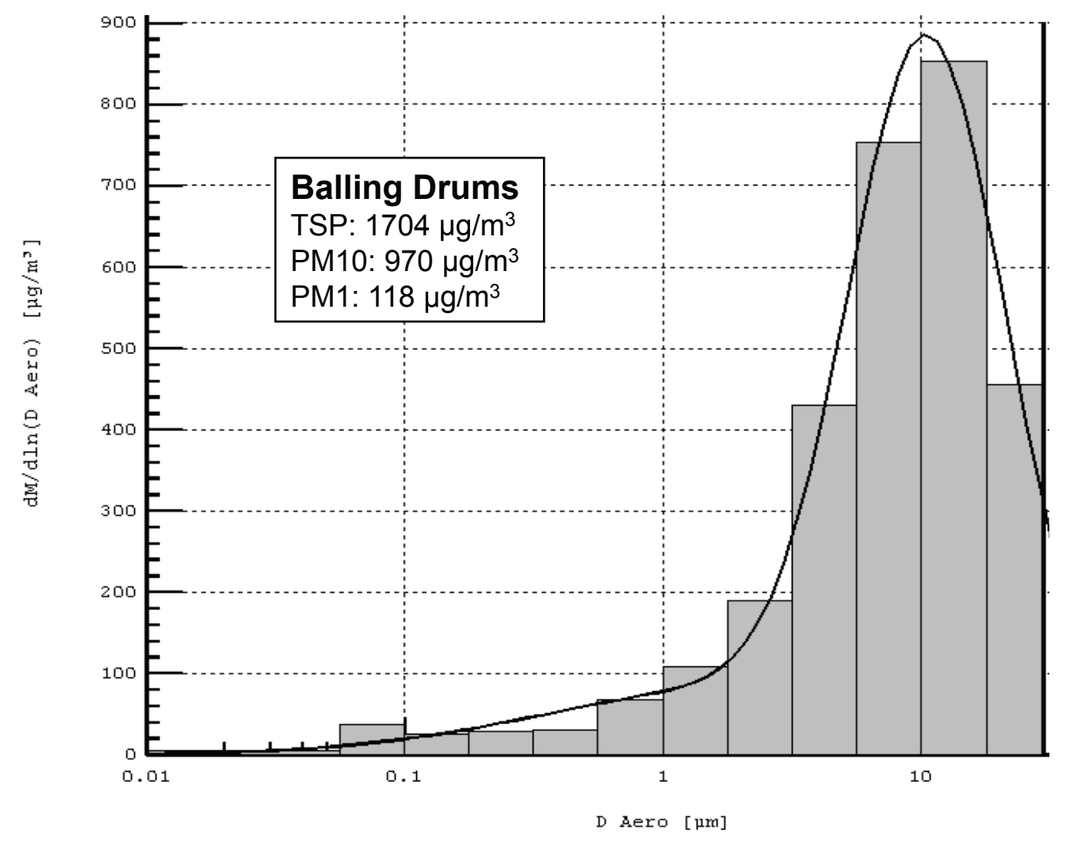
Balling Drums
TSP: 1704 µg/m3
PM10: 970 µg/m3
PM1: 118 µg/m3
dM/dln(D Aero) [µg/m3]
D Aero [µm]

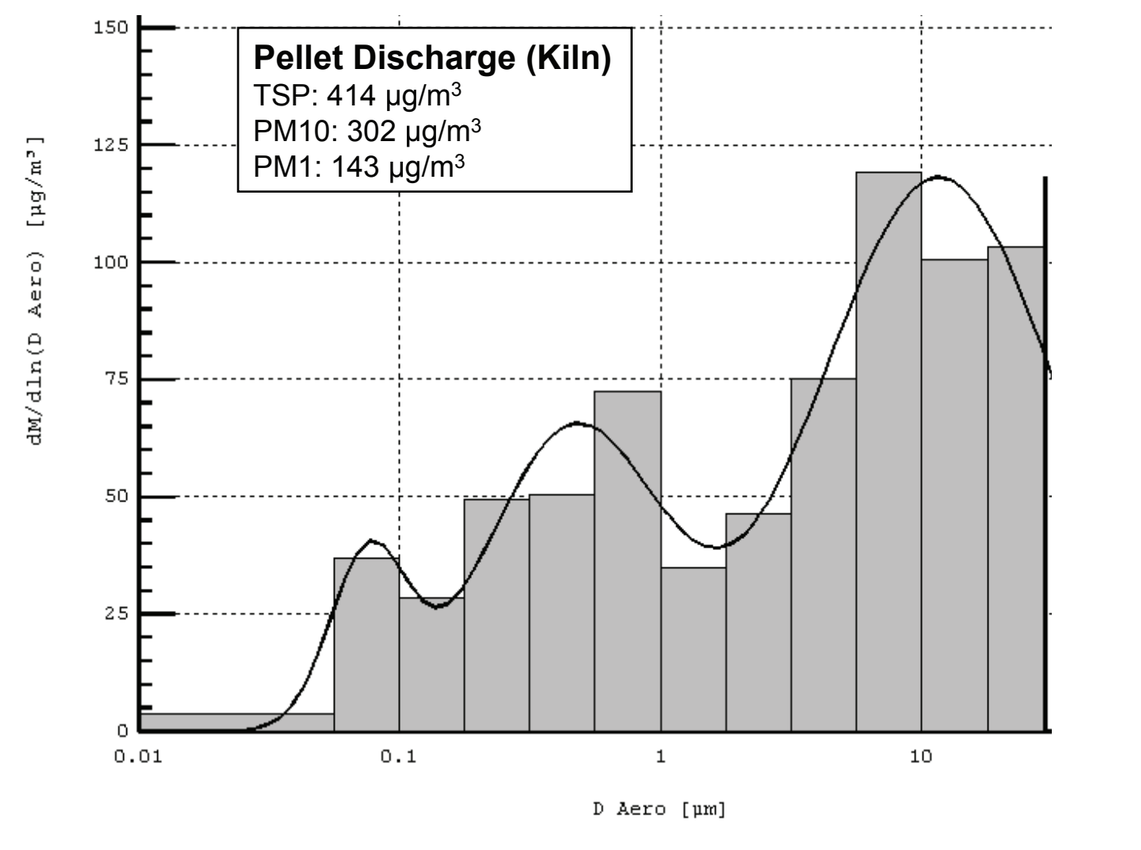
Pellet Discharge (Kiln)
TSP: 414 µg/m3
PM10: 302 µg/m3
PM1: 143 µg/m3
dM/dln(D Aero) [µg/m3]
D Aero [µm]

Simultaneous community sampling data

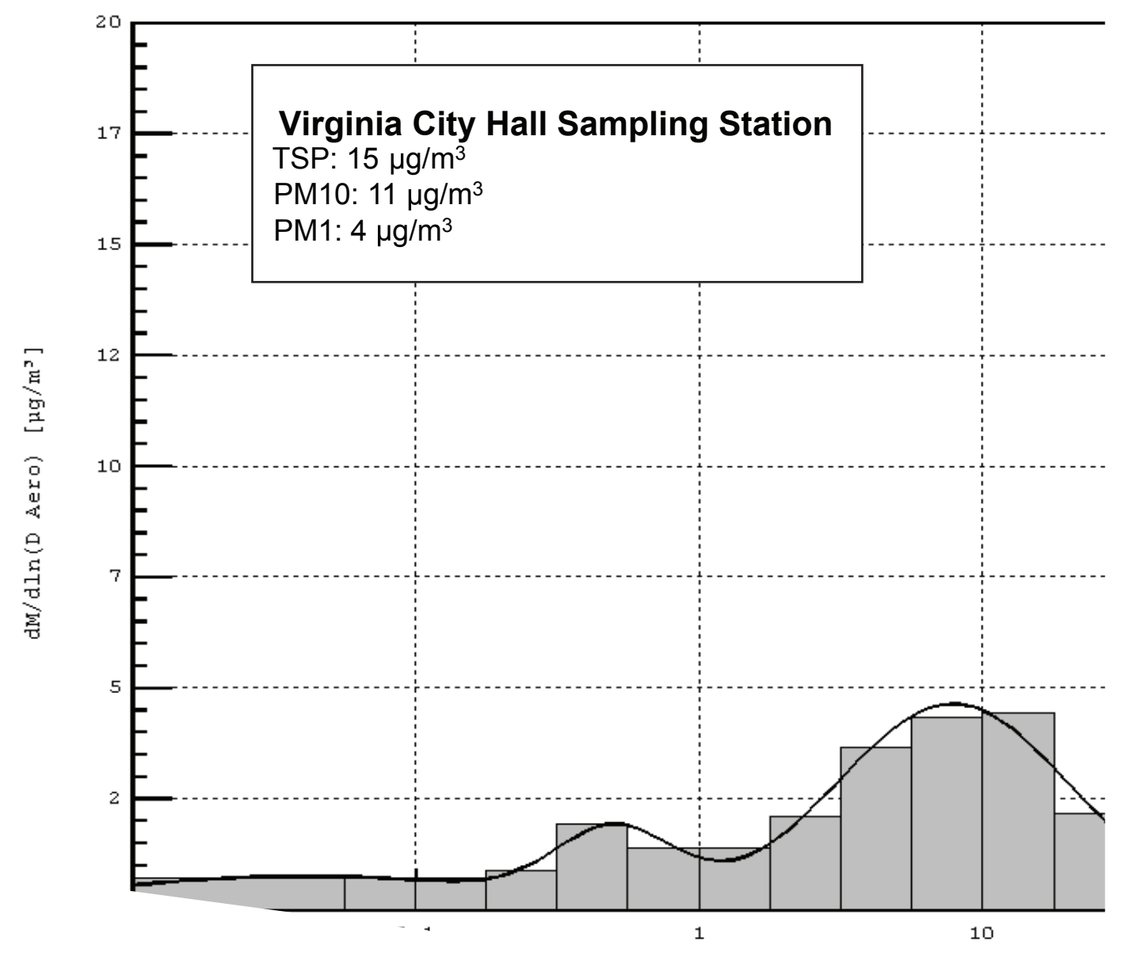
Virginia City Hall Sampling Station
TSP: 15 µg/m3
PM10: 11 µg/m3
PM1: 4 µg/m3
dM/dln(D Aero) [µg/m3]

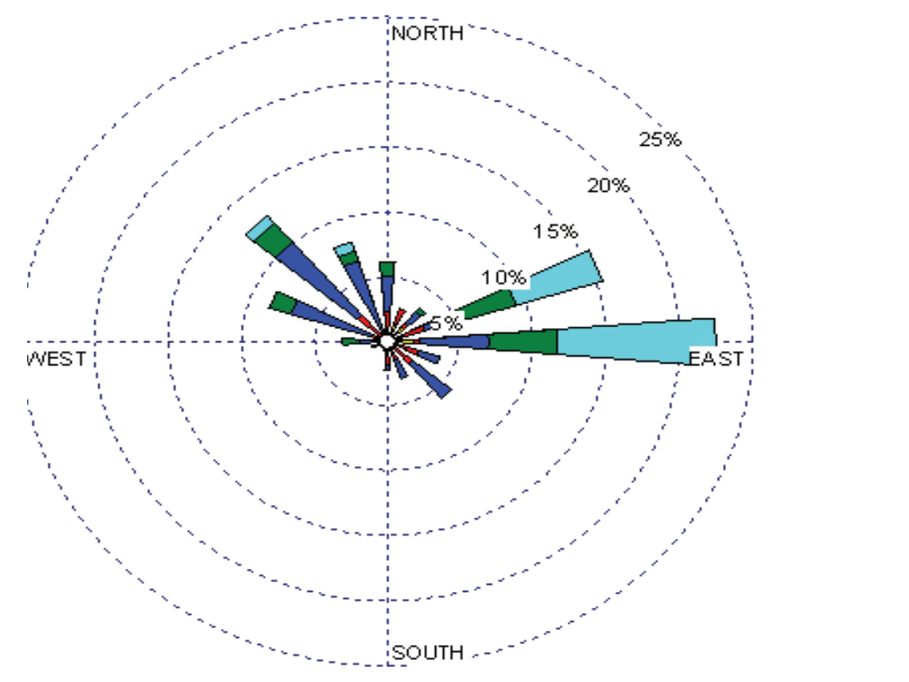
NORTH
SOUTH
WEST
EAST
25%
20%
15%
10%
5%

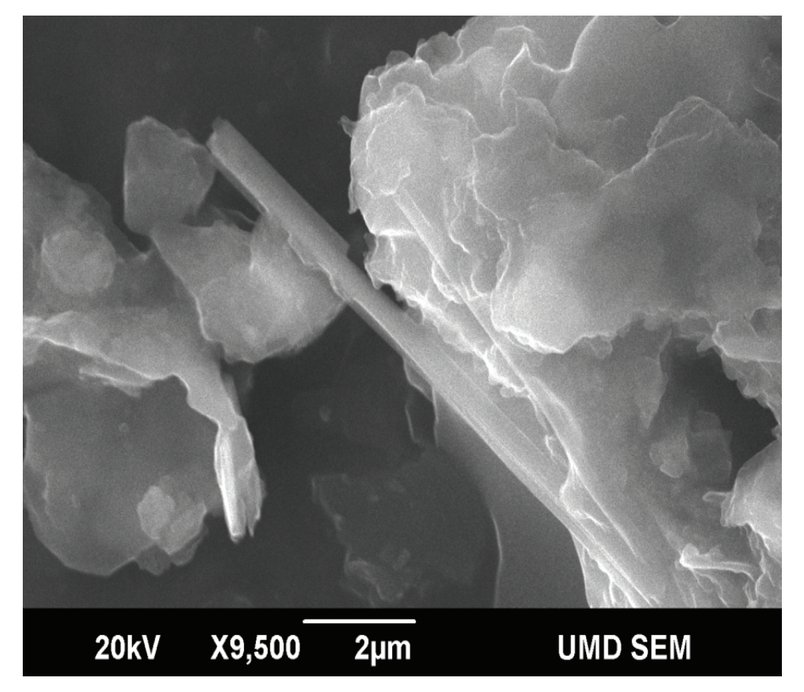
20kV X9,500 2µm UMD SEM

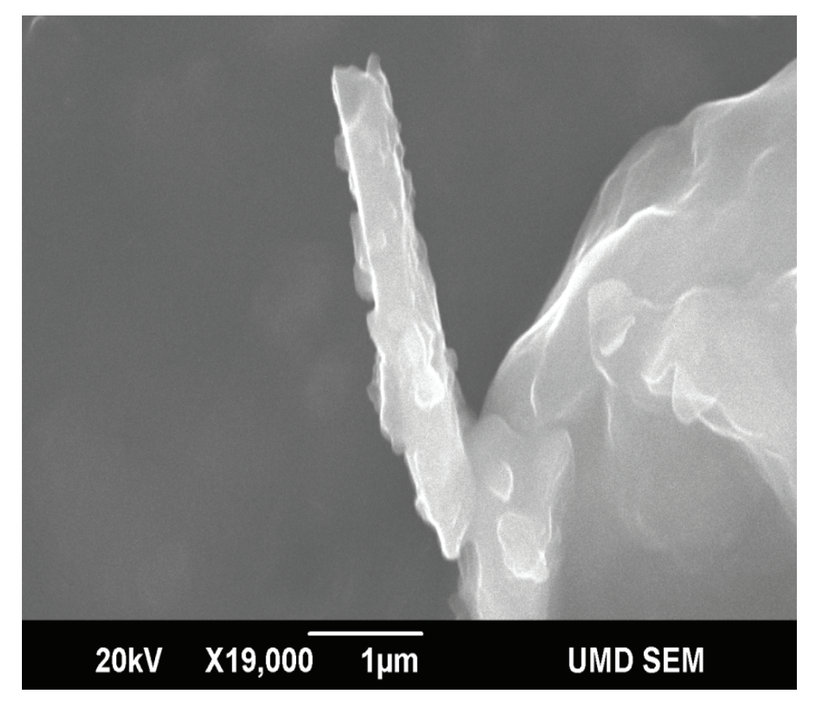
20kV X19,000 1µm UMD SEM

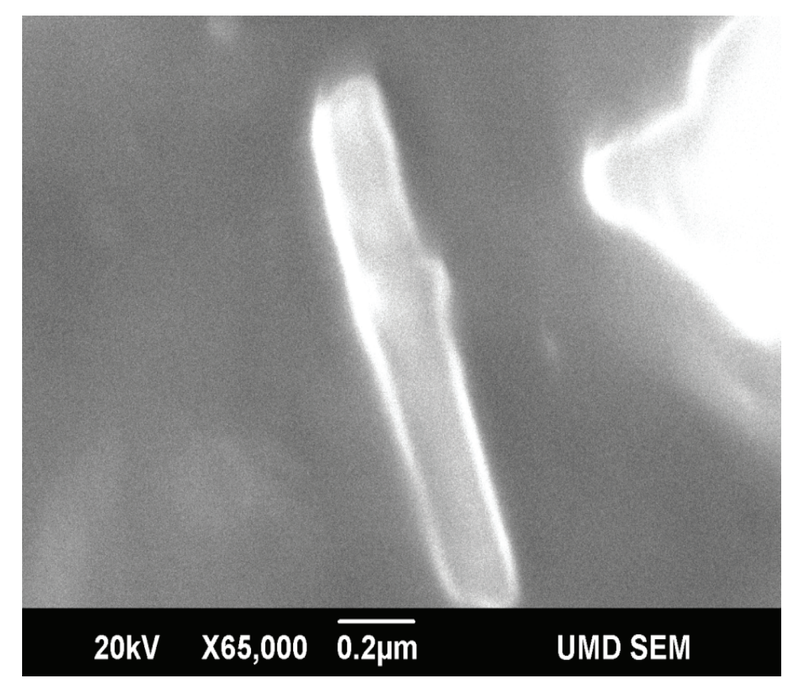
20kV X65,000 0.2µm UMD SEM

Scanning Electron Images of particles collected at the Virginia City Hall Sampling Station.

FUTURE WORK

By far, the majority of the work on this project is ahead of us. Regular sample collection will continue through October 2010, with targeted sampling being conducted for approximately one additional year. Analysis will be ongoing through approximately October 2011. In addition to the gravimetric analysis and electron microscopy, we will be conducting XRD and XRF on the samples.

Taconite Particulate Project Timeline

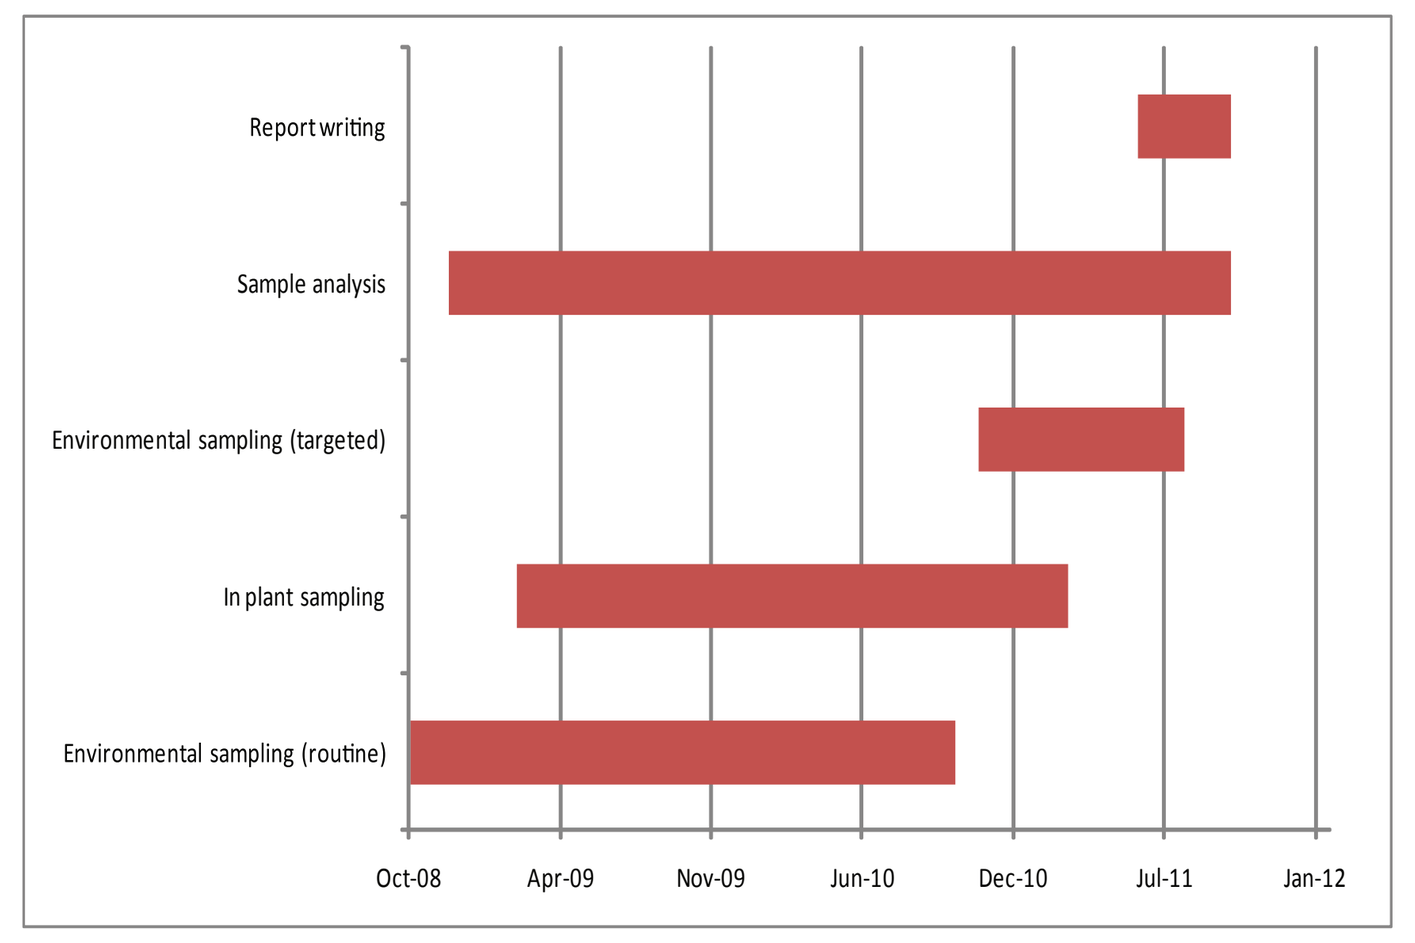
Report writing
Sample analysis
Environmental sampling (targeted)
In plant sampling
Environmental sampling (routine)
Oct-08
Apr-09
Nov-09
Jun-10
Dec-10
Jul-11
Jan-12

Every reasonable effort has been made to ensure the accuracy of the factual data of which this map interpretation is based; however, the University of Minnesota does not warrant or guarantee that there are no errors. Users may wish to verify critical information. In addition, every effort has been made to ensure that the interpretation conforms to sound geologic and cartographic principles, however, no claim is made that the interpretation is rigorously correct.
© 2009 by the Regents of the University of Minnesota
All rights reserved

The University of Minnesota is an equal opportunity educator and employer.

Authors: T. Diedrich, D. Brecke, M. Schreiber, and L. Zanko
Date: 5/5/2009